A RANDOMIZED TRIAL TO DETERMINE THE OPTIMAL CIRCADIAN TIME OF VINORELBINE ADMINISTRATION COMBINED WITH CHRONOMODULATED INFUSION OF 5-FU IN PREVIOUSLY TREATED PATIENTS WITH METASTATIC BREAST CANCER EORTC STUDY 05971

EORTC European Organisation for Research and Treatment of Cancer

http://www.eortc.be

B. Coudert[1], C. Focan[2], N. Tubiana[3], S. Giacchetti[4], F. Lévi[5], F. Cvickovic[6], A. Zambelli[7], G. Fillet[8], P. Chollet[9],M.A. Lentz[10], S. Marreaud[10], B. Baron[10], T. Gorlia[10]

[1]Centre Georges-François-Leclerc, Dijon, France; [2]Centre Hospitalier Saint-Joseph, Liege, Belgium ; [3]C.H.R.U. de Limoges , Limoges, France ; [4]Hopital Saint-Louis, Paris, France ; [5]Hopital Paul Brousse, Paris, France ; [6] Centre René Hugenin, Saint-Cloud, France; [7]Fondazione Salvatore Maugeri, Pavia, Italy; [8]C.H.U. Sart-Tilman, Liege, Belgium; [9]Centre Jean Perrin , Clermond- Ferrant, France; [10]EORTC Data Center, Brussels, Belgium.

Supported by Pierre Fabre Oncology, Ligue Bourguignonne contre le Cancer

BACKGROUND

Metastatic breast cancer remains an incurable disease and the treatments aim at relieving symptoms without toxicity and prolonging survival. The association of 5-FU and vinorelbine is one of the most utilized chemotherapy scheme, but hematologic toxicity remains a problem. Chronotherapeutics aim at determining the most active treatment schedule, based on the adjustment of drug delivery to the circadian timing to reduce toxicities for host cells and enhance efficacy against cancer cells. The EORTC Chronotherapy Study Group has developed an optimal chronomodulated delivery scheme of 5-FU with less toxicity and better efficacy, between 22.00 (10 pm) and 10.00 (10 am) with peak delivery rate at 4.00 (4 am). Experimental data of vinorelbine chronotherapy in animal breast cancer model, (Filipski E et. al. J Pharmacol Exp Ther 1999) show that given at 7 or 11 HALO [4.00 and 8.00 in human subjects] as compared to 19 or 23 HALO [17.00 (5 pm) and 21.00 (9 pm) in human subjects] vinorelbine is more toxic on bone marrow, is more lethal in mice, allows more tumor progression and survival is worse. The aims of the EORTC 05971 trial was to define, in humans, the least toxic time of vinorelbine administration, in a regimen where this drug was combined with a chronomodulated infusion of 5-FU, in patients with metastatic breast cancer.

OBJECTIVES

Definition of the **Least Toxic Time (LTT)** of vinorelbine, given in combination with 5-FU in metastatic breast cancer patients.

Primary endpoint : incidence of G3-4 neutropenia over 3 cycles

Secondary endpoints: incidence of other toxicities, dose intensity of each drug, time of best tolerability (dose reduction, dose delay, treatment interruption for toxicity reason).

PATIENTS & METHODS

Key Eligibility Criteria

- Metastatic breast cancer (MBC)
- Maximum one chemotherapy regimen either for MBC or for adjuvant/neoadjuvant treatment within the last year
- WHO performance status ≤ 2
- Adequate hematologic, hepatic, renal and cardiac function
- No central nervous system metastasis

Treatment regimen

3 courses of chemotherapy (D1-D6) Q3 wks

- D1 & D6 vinorelbine at a fixed dose of 30 mg/m^2/d, 20 minutes I.V infusion
- D2-D5 5-FU at a fixed dose of 850 mg/m^2/d, I.V infusion, administration according to a sinusoidally-modulated delivery rate from 22:00 to 10:00 with a maximum at 4:00

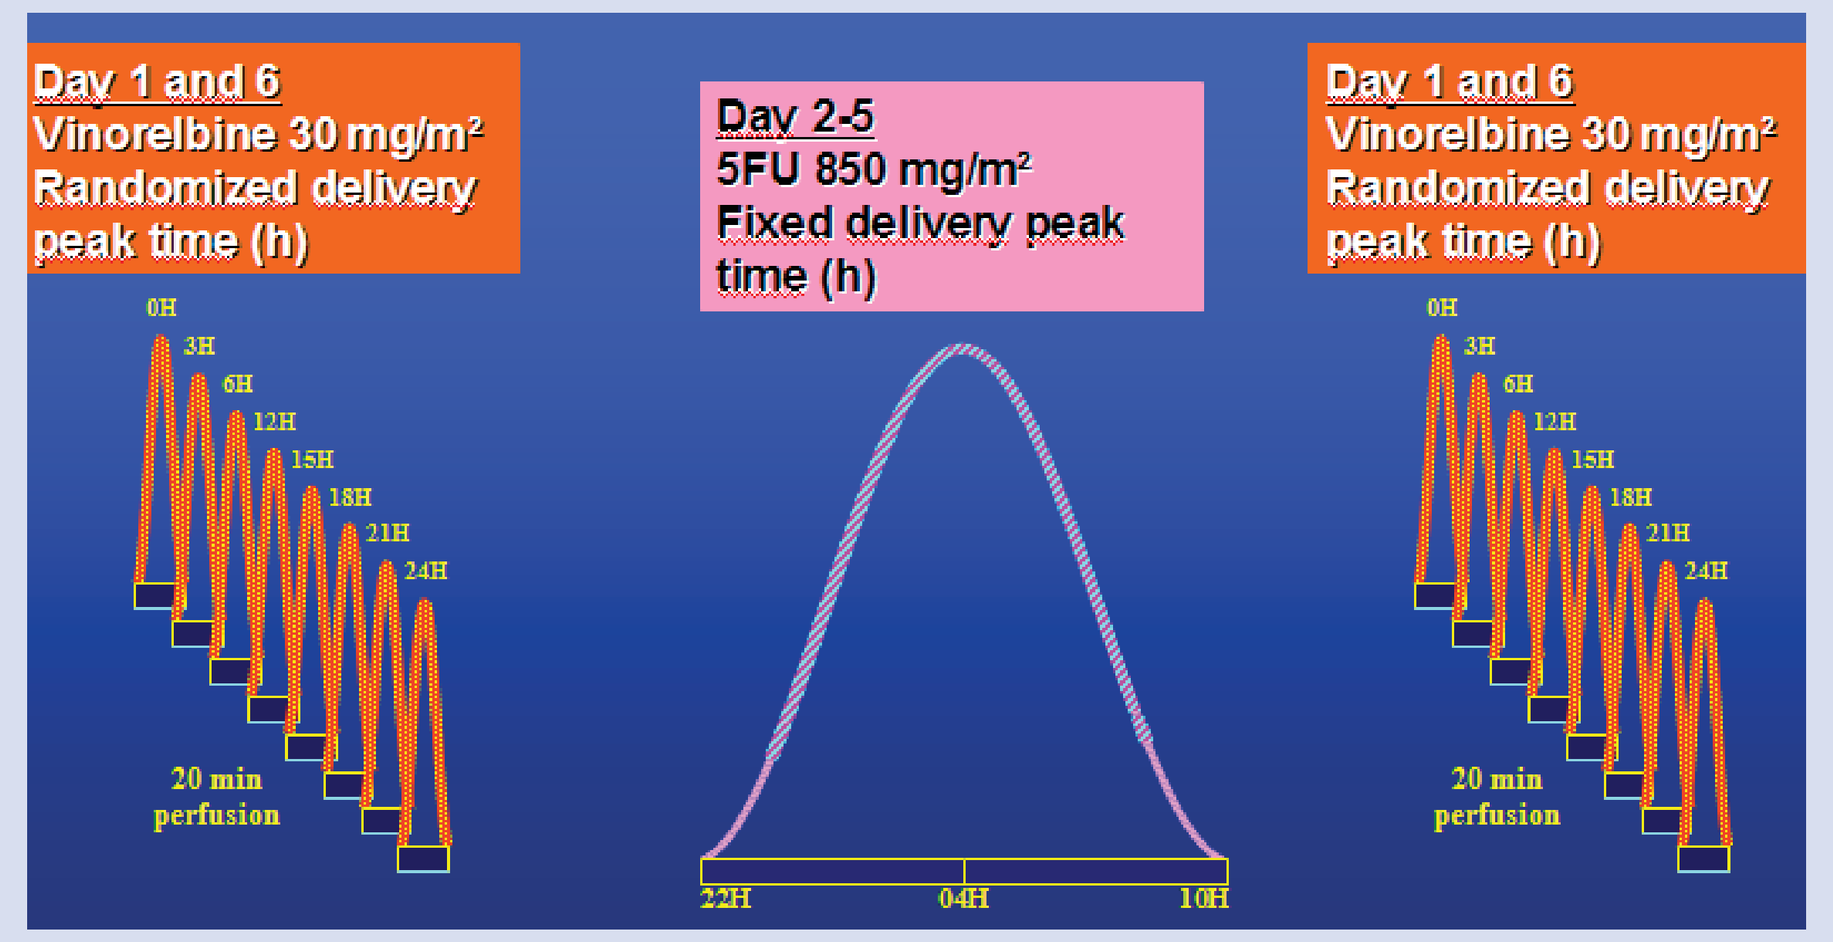
Day 1 and 6
Vinorelbine 30 mg/m²
Randomized delivery
peak time (h)
Day 2-5
5FU 850 mg/m²
Fixed delivery peak
time (h)
Day 1 and 6
Vinorelbine 30 mg/m²
Randomized delivery
peak time (h)
20 min perfusion
22H
04H
10H
20 min perfusion

Safety assessment

Toxicity was assessed according to the NCIC-CTCAE version 2.0

Dose reduction were planned for the following toxicities:
G3 constipation, G1-2 neuropathy, G3-4 leucopenia, neutropenia or thrombocytopenia, febrile neutropenia, G3-4 mucositis or diarrhea, G3-4 vomiting (if uncontrolled with setrons), other G3 toxic symptom discussed on case by case.

Occurence of Toxicity	Dose Reduction	
	5-FU (mg/m²/d)	Vinorelbine (mg/m²/d)
First episod	Reduction to 650	Reduction to 25
Second episod	Reduction to 500	Reduction to 20

An Independent Data Monitoring Committee was created to give advices on the study development.

Statistics

The Least Toxic Time (LTT) was estimated using a logistic regression model assuming a sinusoidal distribution over time (i.e. over the 8 different arms) of the toxicity rate observed in each arm. The associated 90% confidence limits (CL) was obtained by bootstrap method. Parametric methods for the estimation of the CL was used to check the consistency of the results. Only 90% CI of less than 6 hours width was considered as significant and could be implemented in clinical practice.

The multicenter randomised trial used 8 groups of 10 patients each. Vinorelbine was assigned to each group at one of the 8 dosing times: 3h apart (00:00, 03:00, 06:00, 09:00, 12:00, 15:00, 18:00 or 21:00). Patients were stratified by centre and previous chemotherapy (2nd vs. 3rd line) and randomised into one of the 8 groups, using the minimisation technique.

RESULTS

Patient 's characteristics

From 1999 until 2004, 90 patients with MBC were recruited. Seven patients were ineligible, due to cerebral metastasis (1 pt), prior radiotherapy on bone (2 pts), insufficient delay between the last treatment and the inclusion in the study (2 pts) and no treatment after randomization (2 pts), leading to the analysis of 83 patients.

Variable		Total (N=83)
		N (%)
Age	Median	57.8
	Range	30.0 - 1.2
WHO Performance Status	WHO PS0	48 (57.8)
	WHO PS1	32 (38.6)
	WHO PS2	3 (3.6)
Prior surgery	No	9 (10.8)
	Yes, curative	67 (80.7)
	Yes, palliative	7 (8.4)
Prior radiotherapy	No	22 (26.5)
	Yes, primary / axillary / mammary	61 (73.5)
Prior(neo)adjuvant chemotherapy	No	35 (42.2)
	Yes, >1 year adj.<>pall. chemo	35 (42.2)
	Yes, <1 year adj.<>pall. chemo	13 (15.7)
Prior palliative chemotherapy	No	5 (6.0)
	Yes, 1 line	57 (68.7)
	Yes, 2nd lines	21 (25.3)
Prior high dose chemotherapy	No	83 (100)
Prior hormonotherapy	No	23 (27.7)
	Yes	60 (72.3)
Prior immunotherapy or BRM	No	80 (96.4)
	Yes	3 (3.6)

Treatment

After the inclusion of 46 patients, due to toxicity, according to the IDMC recommendations, the dose of vinorelbine was reduced to 25 mg/m²/d: 40 patients were treated with vinorelbine 30 and 43 with vinorelbine 25.

Treatment duration	Percentage of patients
3 cycles	81 %
2 cycles	8 %
1 cycle	11 %

Cycles, with pump technical problems leading to a more than 25% deviation in the drug dosage or timing, were excluded from the final analysis. A total of 224 cycles have been analyzed:

Vinorelbine 30: 40, 33, 27
(first, second and third cycle respectively)

Vinorelbine 25: 43, 41, 40
(first, second and third cycle respectively)

	Dose intensity (%)
Vinorelbine 30 mg/m²/d	79.4
5-FU combined with V30	78.2
Vinorelbine 25 mg/m²/d	88.1
5-FU combined with V25	87.4

Safety

Adverse events observed over the 3 cycles (percentage per patient):

G3-G4 leucopenia (47% & 29%), G3-G4 neutropenia (12% & 77%). G4 febrile neutropenia (34%). G2 thrombocytopenia & anemia (4%).

The main other G3-G4 toxicity were cardiovascular (2% & 2%), lethargy (4%), diarrhea (4% & 1%), vomiting (1% & 1%), stomatitis (12%), constipation (2% & 4%), other gastrointestinal (5% & 1%), infection (4%), sensory (4%), pulmonary (2% & 3%), alopecia (7%).

Least Toxic Time analysis

The Least Toxic Time was estimated for several toxicities, by group of dose and combined doses. Finally stratification by dose analysis was performed. Based on the stratification by dose analysis, 90% CI of less than 6 hours width was observed:

- around 17:00 (5 p.m.) for the incidence of leucopenia G3-4 (Figure 1-A). This time of administration almost corresponds to the least toxic time and to the most efficient time of administration in animal model (19 HALO).
- around 8:00 (8 a.m.) for the incidence of dose reduction for toxicity reason or dose reduction/delay and treatment interruption for toxicity reason (Figure 1-B).

No other 90% CI of less than 6 hours width could be observed for other toxicity endpoints especially for G 3-4 neutropenia the primary endpoint of the study (Figure 1-C).

Least Toxic Time estimation *

Time [90 % CI]	Dose 30	Dose 25	Both dose	Stratification by dose
Leukopenia G3-4	17:36 [10:40-22:37]	17:03 [13:26-20:49]	17:17 [13:58-20:22]	**17:17 [14:04-20:03]**
Neutropenia G3-4	20:02 [07:15-03:07]	24:00 [15:09-12:00]	20:57 [10:08-06:06]	20:57 [10:48-07:03]
Neutropenia G4	19:51 [07:31-01:43]	07:33 [02:17-11:14]	07:40 [22:15-16:54]	07:40 [22:28-15:56]
Hemato G3	11:34 [06:37-16:44]	17:55 [13:39-21:12]	15:17 [10:16-20:28]	15:17 [10:06-19:34]
Hemato G4	19:51 [09:37-03:03]	07:33 [02:33-11:27]	07:40 [21:49-16:11]	07:40 [22:28-15:56]
Febrile Neutropenia	20:13 [15:07-02:02]	06:28 [20:13-14:55]	21:39 [12:31-06:16]	21:39 [12:06-05:58]
Mucositis Diarrhea G3-4	16:08 [13:26-21:07]	09:05 [05:09-12:00]	13:19 [08:49-16:32]	13:19 [09:24-16:40]
Gastrointestinal G3-4	09:55 [06:56-12:03]	14:14 [04:22-20:30]	10:42 [07:15-14:50]	10:27 [06:58-14:15]
Neuropathy G1-2	01:46 [23:20-04:45]	12:00 [03:10-00:16]	01:54 [22:12-07:13]	01:54 [22:29-06:20]
Dose Reduction by Toxicity	07:25 [23:37-12:59]	07:25 [05:34-09:45]	07:27 [05:00-09:52]	**07:27 [05:24-09:53]**
Tolerability **	07:30 [19:53-17:41]	08:09 [06:29-10:28]	08:16 [05:38-11:05]	**08:16 [06:04-10:39]**

*The least toxic time (LTT) was estimated using a logistic regression model assuming a sinusoidal distribution over time (i.e. over the 8 different arms) of the toxicity rate observed in each arm. The associated 90% confidence limits (CL) was obtained by bootstrap method (REF 1: Baron et al. 2001 /ID550). Parametric methods for the estimation of the CL was used to check the consistency of the results. Only a 90% CI of less than 6 hours width was considered as significant (in bold in the table)

**Tolerability was defined as the time of administration with less dose reduction, dose delay, treatment interruption for toxicity reason

The optimal time for 'strat by dose' is
17:17 (90 CI: 14:04 to 20:03)

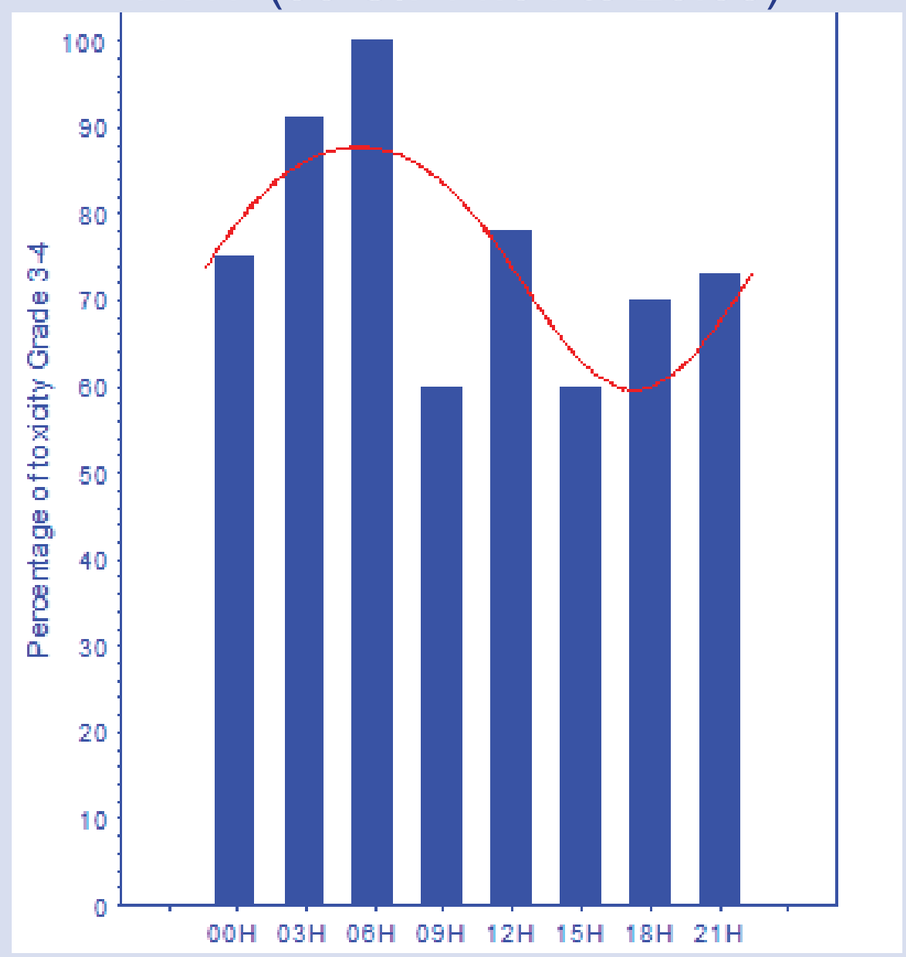
Percentage of toxicity Grade 3-4
00H 03H 06H 09H 12H 15H 18H 21H

A. Leucopenia G3-G4

The optimal time for 'strat by dose' is
8:16 (90 CI: 6:04 to 10:39)

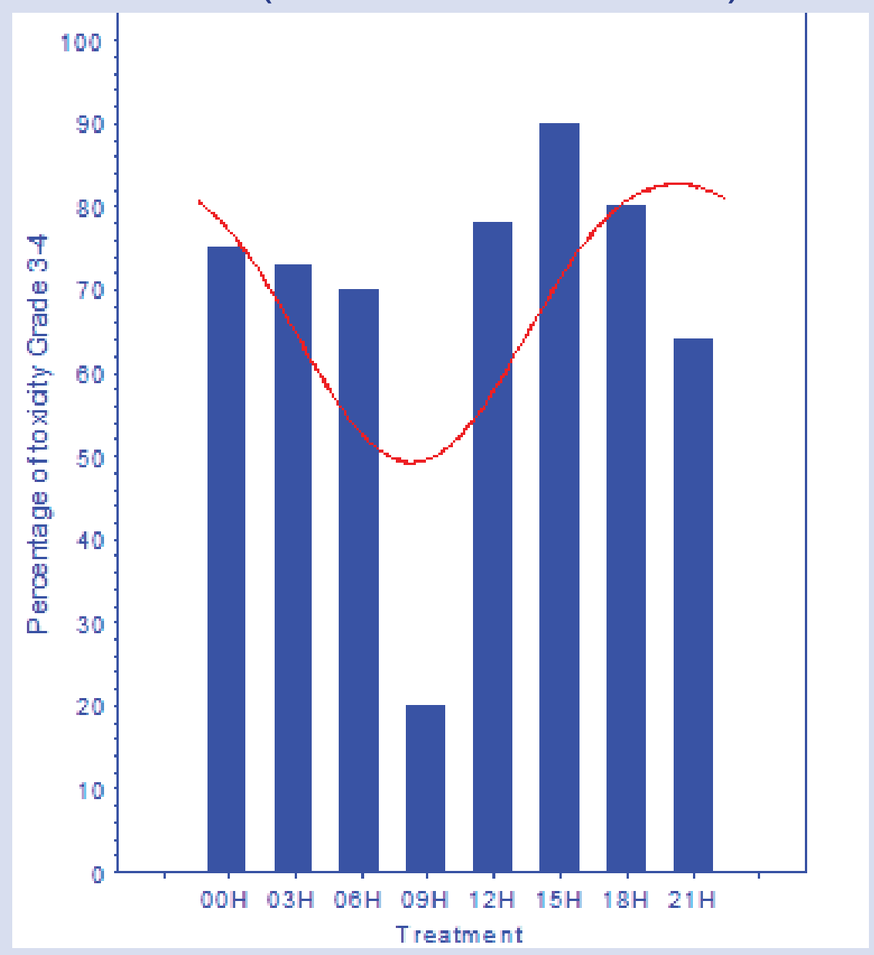
Percentage of toxicity Grade 3-4
00H 03H 06H 09H 12H 15H 18H 21H
Treatment

B. Tolerability

The optimal time for 'strat by dose' is
20:57 (90 CI: 10:48 to 7:03)

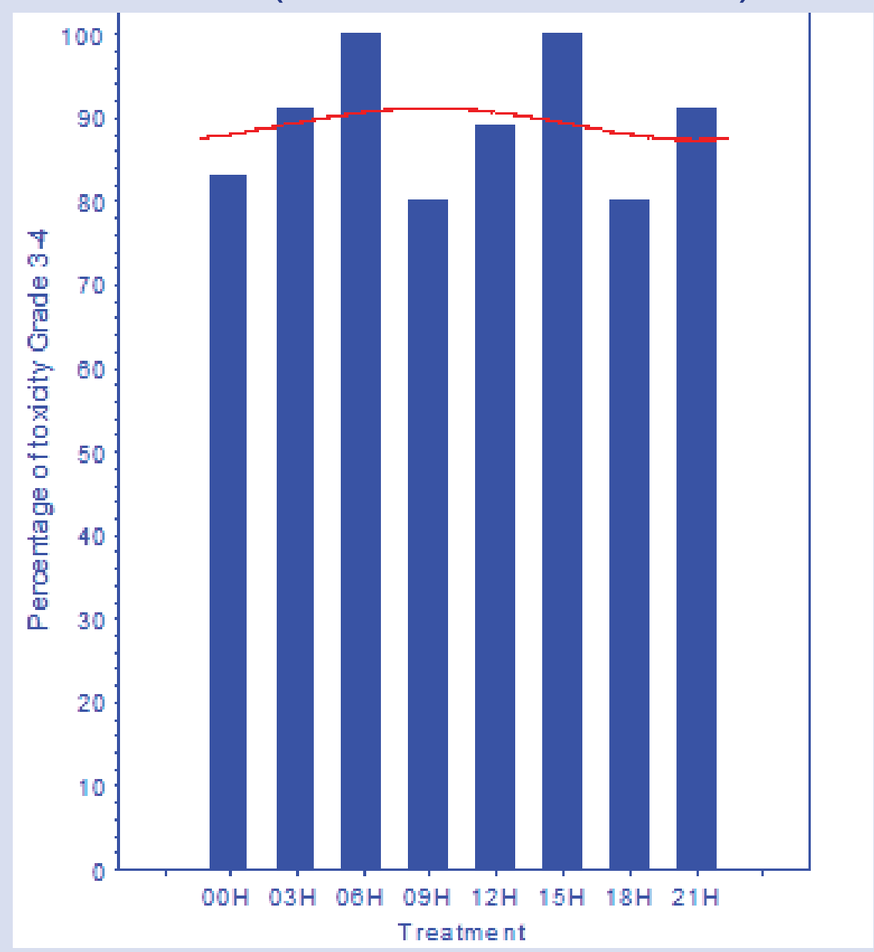
Percentage of toxicity Grade 3-4
00H 03H 06H 09H 12H 15H 18H 21H
Treatment

C. Neutropenia G3-G4

CONCLUSION

Chronomodulation of vinorelbine in Humans can demonstrate an optimal time of administration at 17:00 (5 p.m.) to reduce the incidence of leucopenia G3-4. This time of administration almost corresponds to the least toxic time and to the most efficient time of administration in animal model (19 HALO). This is the first time that a chronobiological animal pre-clinical data and a logistic regression with bootstrap method statistical design are reinforced by a randomized clinical studies in Humans.

Neutropenia G3-4, the main endpoint of this study was not modified by the time of administration of vinorelbine. The main reason for this observation was that neutropenia G3-4 was almost above 80%, at all times of vinorelbine administration. Indeed, due to its frequency, the toxicity lead to the reduction of the vinorelbine dosage after the inclusion of 46 patients. Tolerability, a more general concept in drug delivery must be tested in other studies.